61ST ERA CONGRESS
STOCKHOLM & VIRTUAL
MAY 23-26, 2024

Oral Administration of UNI-494 Ameliorates Acute Kidney Injury in a Rat Model of Delayed Graft Function

S. MEDICHERLA[1]; G. REDDY[1]; S GUPTA[1]

[1] Unicycive Therapeutics, Inc.

UNICYCIVE THERAPEUTICS INC.

BACKGROUND

- There are no FDA approved drugs for the treatment of acute kidney injury (AKI), which affects 10-15% of hospitalized patients and often results in renal transplantation or lifelong dialysis
- Inflammation and reactive oxygen species (ROS) driven mitochondrial permeability transition pore (mPTP) opening causes mitochondrial dysfunction/swelling and eventual cell death over time
- This is implicated in a wide range of acute diseases including acute kidney disease (AKI) originating from ischemia reperfusion injury (IRI) or delayed graft function (DGF)
- Furthermore, unresolved inflammation exacerbates sustained mPTP opening, evident in chronic kidney diseases (CKD)
- Nicotinamide adenine dinucleotide (NAD^+) can suppress the frequency and duration of mPTP opening
- UNI-494 is a novel nicotinamide ester derivative that is a selective mitochondrial ATP-sensitive potassium channel (KATP) activator, which reverses the mitochondrial dysfunction by closing the mPTP
- UNI-494 may be beneficial for several disease states, including AKI

OBJECTIVE

We present results from a study evaluating the in vivo efficacy of oral (PO) UNI-494 to prevent damage in the unilateral renal ischemia-reperfusion (I/R) rat model of AKI, which is a well-established model of DGF[1]

METHODS

- Rats were anesthetized, the right kidney was removed, and I/R was induced in three groups of rats by clamping the renal vessels in the left kidney for 30 minutes; a fourth group had no surgery (sham group) without clamping the renal vessels
- 50 mg/kg or 100 mg/kg UNI-494 were administered PO 30 minutes prior to I/R **(Figure 1)**
- One I/R group and the sham group were not treated
- After 24 hours of reperfusion in metabolic cages, blood samples were collected for serum creatinine (sCr) and BUN
- 24-hour urine samples were collected for albumin-creatinine ratio (ACR) and the tubular injury marker neutrophil gelatinase-associated lipocalin (NGAL)
- At necropsy, the clamped left kidney was collected and processed for histology for proximal tubular injury scores

Figure 1. UNI-494 I/R Study Design – Preventive Dosing (PO)

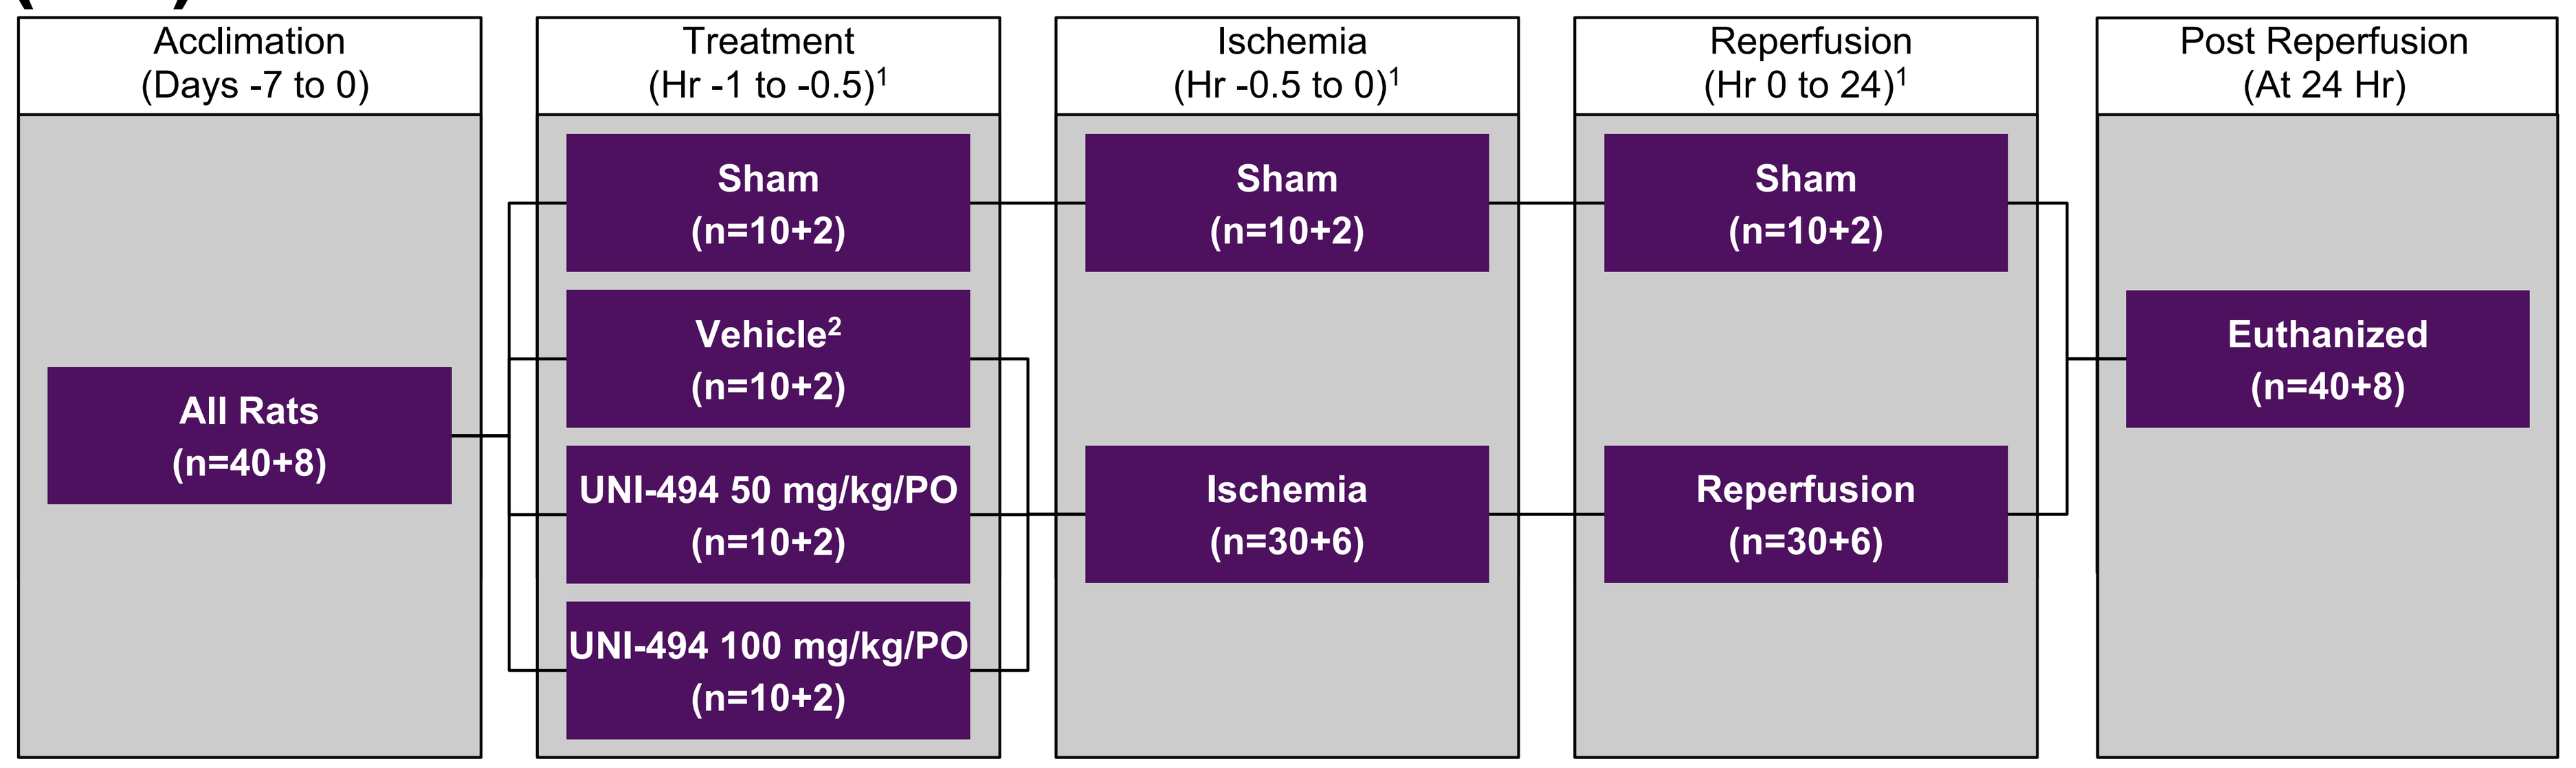
Acclimation (Days -7 to 0)
Treatment (Hr -1 to -0.5)1
Ischemia (Hr -0.5 to 0)1
Reperfusion (Hr 0 to 24)1
Post Reperfusion (At 24 Hr)
All Rats (n=40+8)
Sham (n=10+2)
Vehicle2 (n=10+2)
UNI-494 50 mg/kg/PO (n=10+2)
UNI-494 100 mg/kg/PO (n=10+2)
Sham (n=10+2)
Ischemia (n=30+6)
Sham (n=10+2)
Reperfusion (n=30+6)
Euthanized (n=40+8)

1 Hr 0 is defined as the start of reperfusion
2 Vehicle = 0 mg/kg
Note: N number consists of experimental + spare animals
Abbreviations: PO = Oral Gavage

RESULTS

- In this study, I/R induced significant increases of sCr, ACR, NGAL, and proximal tubular injury scores in the vehicle treated I/R group vs No I/R sham group
- A single PO dose of UNI-494 at 50 mg/kg/PO or 100 mg/kg/PO significantly reduced the kidney functional markers (sCr and uACR), the tubular injury marker uNGAL, and proximal tubular injury scores **(Figure 2 & 3)**
- All rats survived to the scheduled euthanasia time point. No clinical observations were noted during any phases of the study

Figure 2. Mean (SE) UNI-494 + I/R vs I/R vs No I/R Sham – Serum Creatinine, Serum BUN, Urinary ACR, and Urinary NGAL

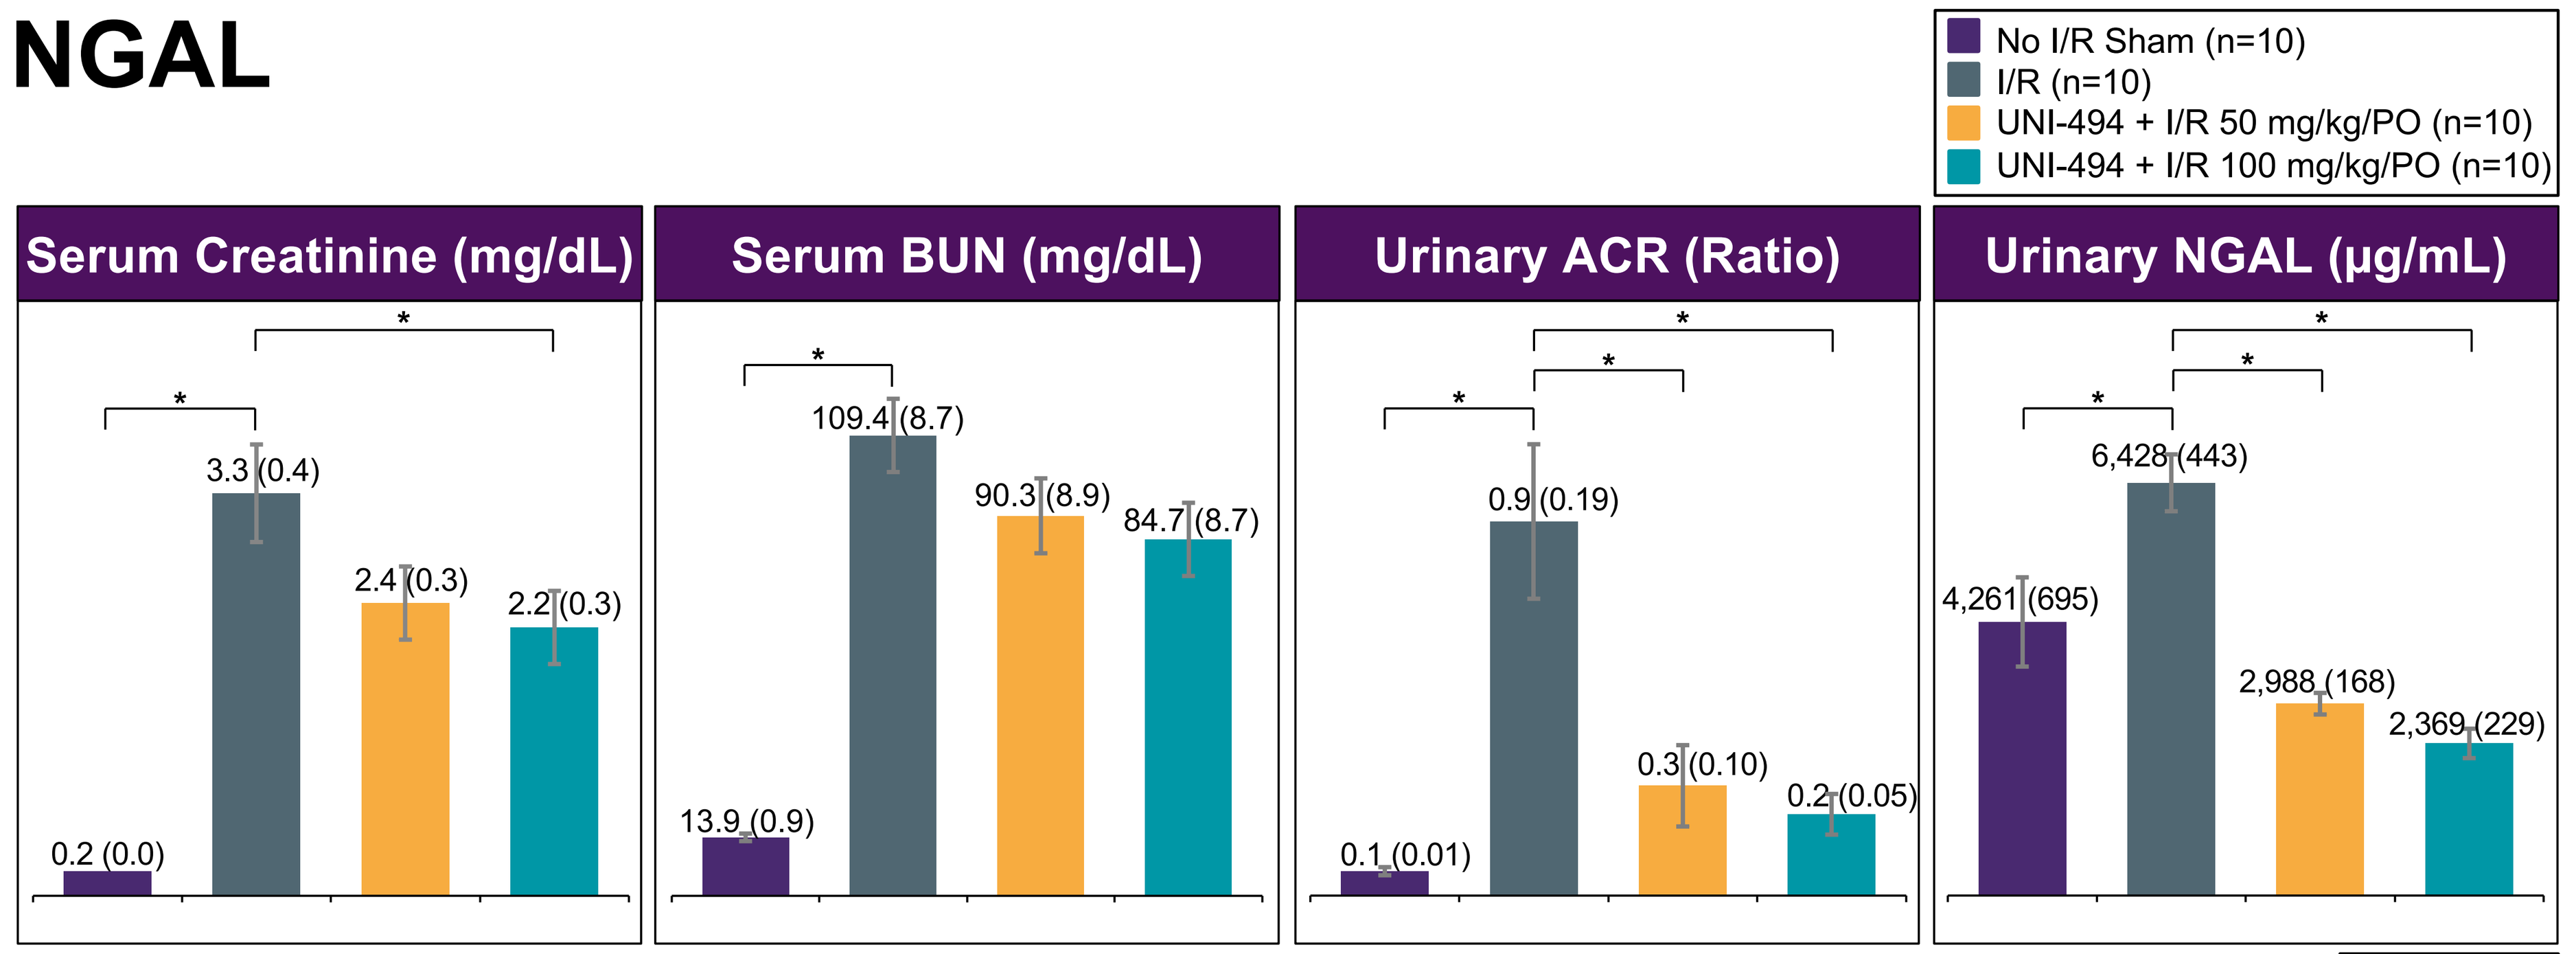
No I/R Sham (n=10)
I/R (n=10)
UNI-494 + I/R 50 mg/kg/PO (n=10)
UNI-494 + I/R 100 mg/kg/PO (n=10)
Serum Creatinine (mg/dL)
0.2 (0.0)
3.3 (0.4)
2.4 (0.3)
2.2 (0.3)
Serum BUN (mg/dL)
13.9 (0.9)
109.4 (8.7)
90.3 (8.9)
84.7 (8.7)
Urinary ACR (Ratio)
0.1 (0.01)
0.9 (0.19)
0.3 (0.10)
0.2 (0.05)
Urinary NGAL (µg/mL)
4,261 (695)
6,428 (443)
2,988 (168)
2,369 (229)
*

1 Statistical comparisons were conducted using one-way ANOVA with Dunnett's Multiple Comparison Test
*p<0.05[1]
Abbreviations: I/R = Ischemia-Reperfusion; PO = Oral Gavage; BUN = Blood Urea Nitrogen; ACR = Albumin: Creatinine Ratio; NGAL = Neutrophil Gelatinase-Associated Lipocalin

Figure 3. Histological Image Where the Nature of the Proximal Tubule Injury Is Pointed with Arrows

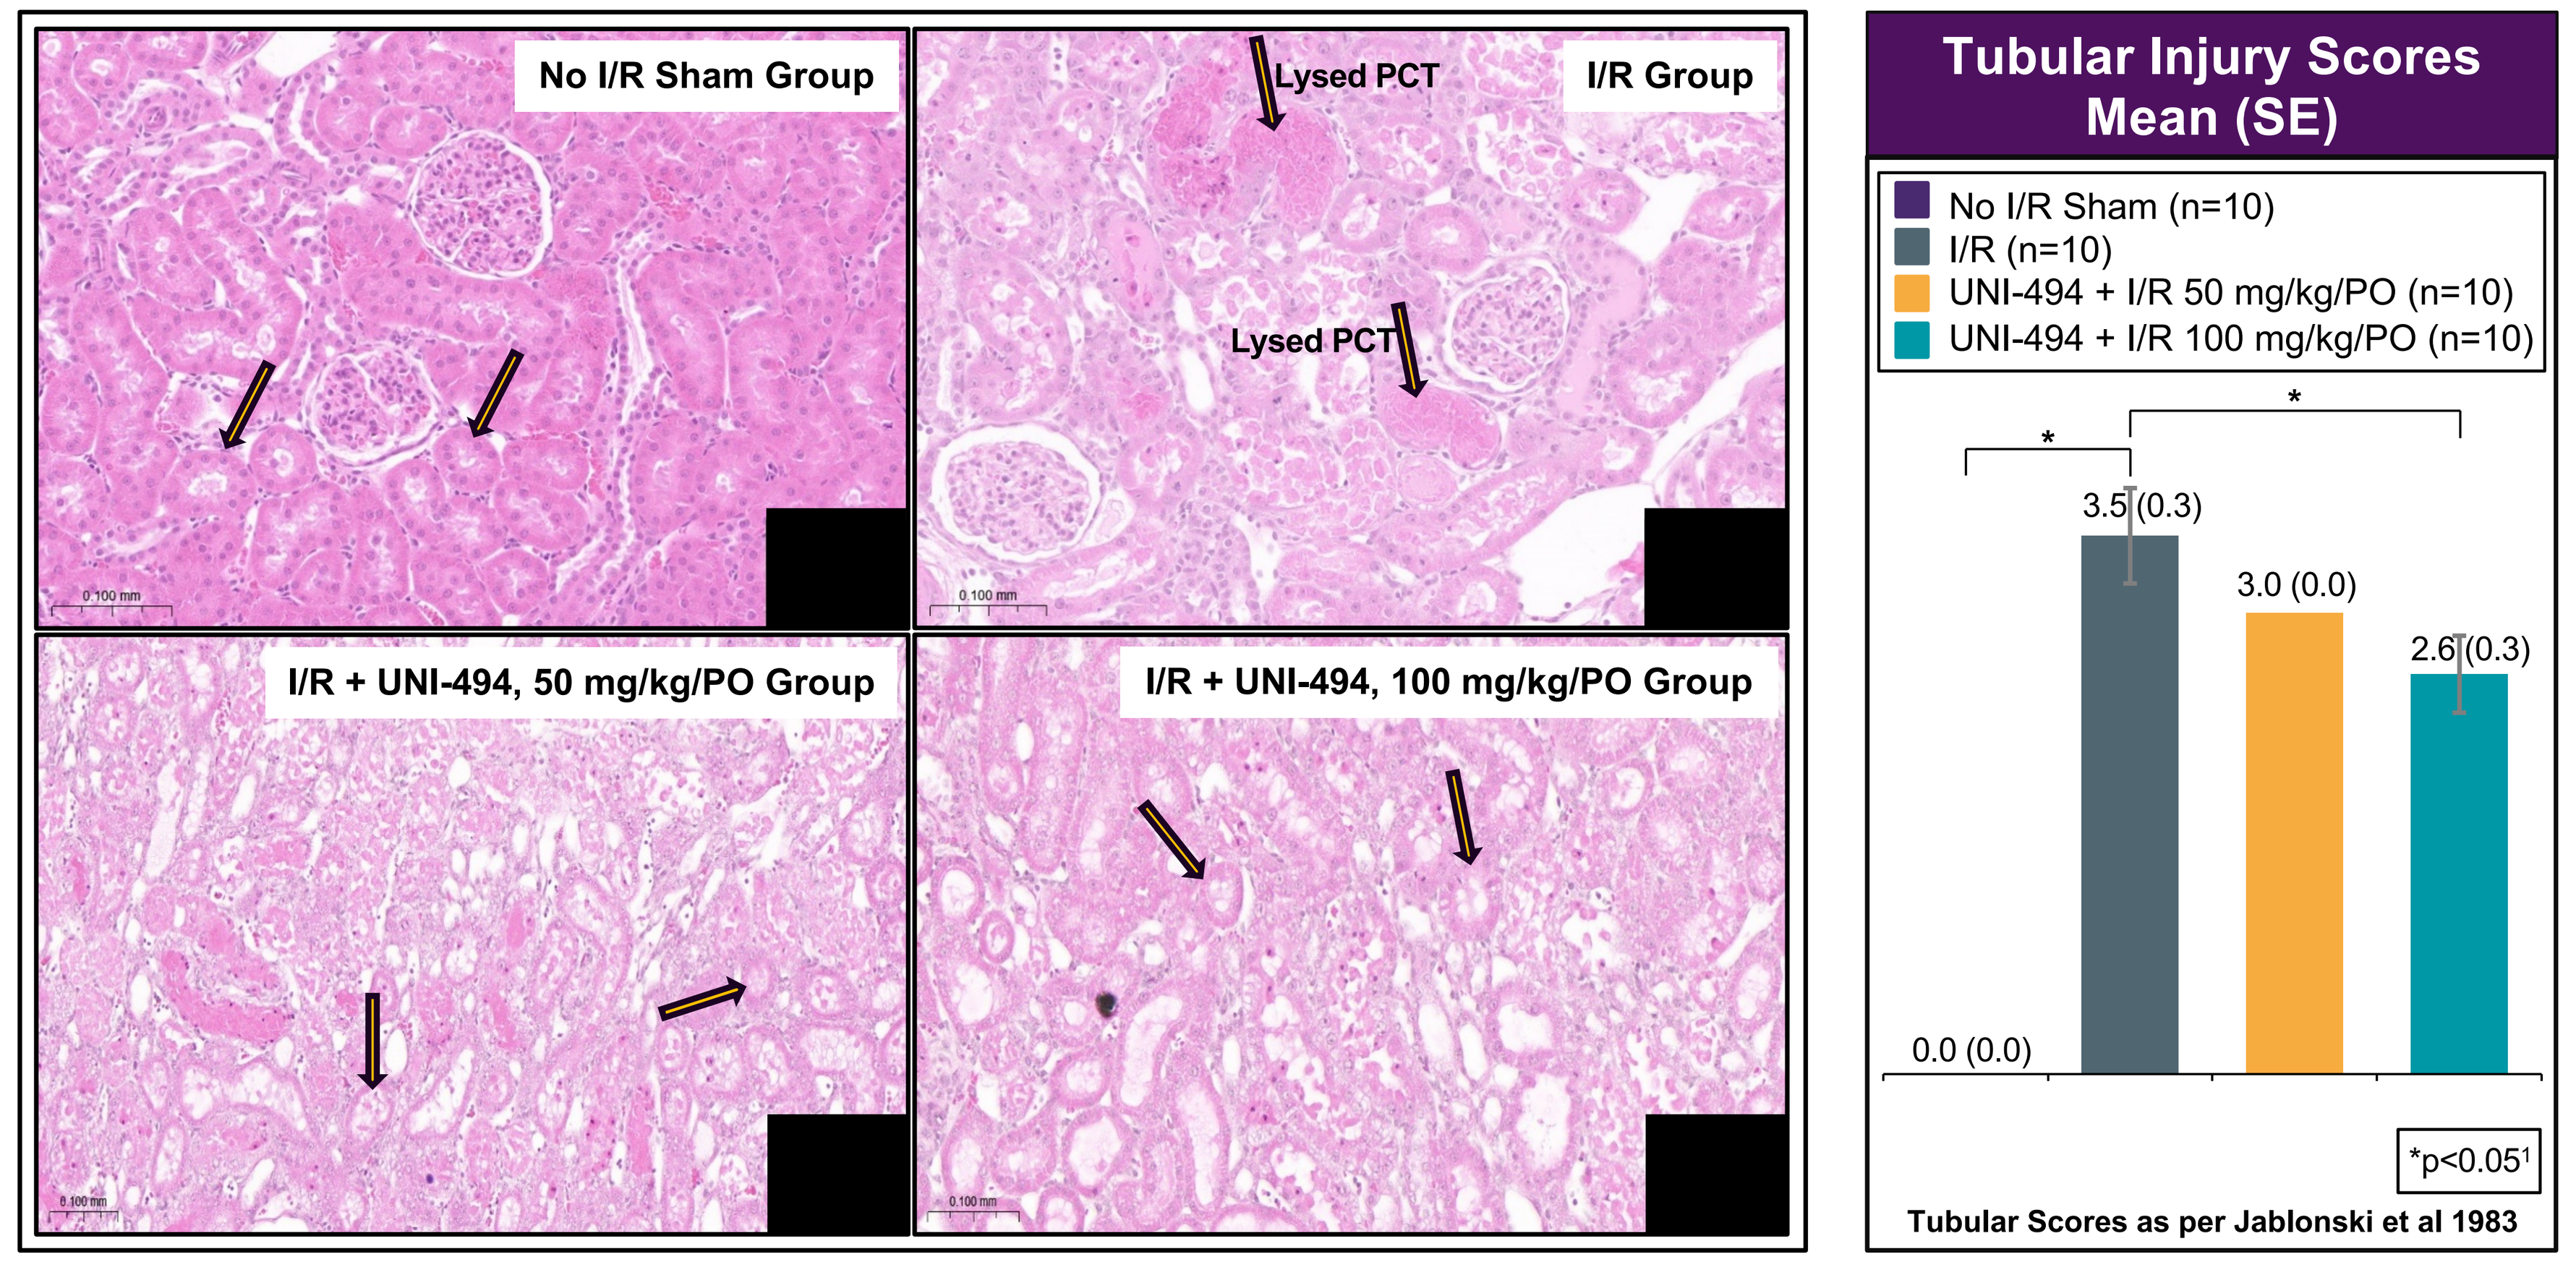
No I/R Sham Group
Lysed PCT
I/R Group
Lysed PCT
I/R + UNI-494, 50 mg/kg/PO Group
I/R + UNI-494, 100 mg/kg/PO Group
Tubular Injury Scores Mean (SE)
No I/R Sham (n=10)
I/R (n=10)
UNI-494 + I/R 50 mg/kg/PO (n=10)
UNI-494 + I/R 100 mg/kg/PO (n=10)
0.0 (0.0)
3.5 (0.3)
3.0 (0.0)
2.6 (0.3)
*
*p<0.05¹
Tubular Scores as per Jablonski et al 1983

1 Dunnett's multiple comparison test was conducted to determine statistical significance while comparing proximal tubular injury scores of other groups with ischemia reperfusion group
Abbreviations: I/R = Ischemia-Reperfusion; PO = Oral Gavage; PCT = Proximal Convoluted Tubule

CONCLUSIONS

- The results indicate that UNI-494 prevented AKI markers and proximal tubular injury scores in a rat model of AKI
- Additionally, kidney functional data (sCR and uACR) were very well supported by lower tubular injury marker uNGAL and proximal tubular injury scores from histology at both low and high doses reflecting a non-dose related trend with biomarkers and is dose-dependent for proximal tubule injury scores
- UNI-494 is a potential candidate for the treatment of patients with DGF and other clinical conditions dealt with AKI

References:
1. Cavaillé-Coll M. et al., *Am J Transplant*. 2013. May.

Acknowledgments:
Writing support was provided by Xelay Acumen Group, Inc., and funded by Unicycive Therapeutics, Inc.

Presented at the ERA Annual Meeting 2024, May 23-26, 2024, Stockholm, Sweden